Targeted *in vivo* generation of CAR T and NK cells utilizing an engineered lentiviral vector platform

James I. Andorko[1], Saar Gill[2,3], and Philip R. Johnson[1]

[1]Interius BioTherapeutics, Inc., Philadelphia, PA [2]Department of Medicine, Division of Hematology-Oncology, University of Pennsylvania Perelman School of Medicine, Philadelphia, PA [3]Center for Cellular Immunotherapies, University of Pennsylvania Perelman School of Medicine, Philadelphia, PA

Proprietary platform for *in vivo* gene delivery

CAR T cell therapies have revolutionized treatment of B cell malignancies. *Ex vivo* CAR T cell manufacturing is complex, costly, and cumbersome, prompting efforts that support shorter manufacturing processes and allogeneic or "universal donor" strategies. Recent data show that CAR T product efficacy is correlated with reduced *ex vivo* manipulation of cells. We have designed a system to transduce target cells *inside the body* to generate autologous CAR cells, circumventing *ex vivo* manipulation, avoiding the need for conditioning chemotherapy, and providing an "off-the-shelf" therapy for B cell malignancies.

INT2104 is a non-replicating, self-inactivating lentiviral vector delivering a transgene for a CAR specific for CD20 (CAR20). The lentiviral vector is designed to generate CAR T and CAR NK cells *in vivo* following a single intravenous administration of the viral vector.

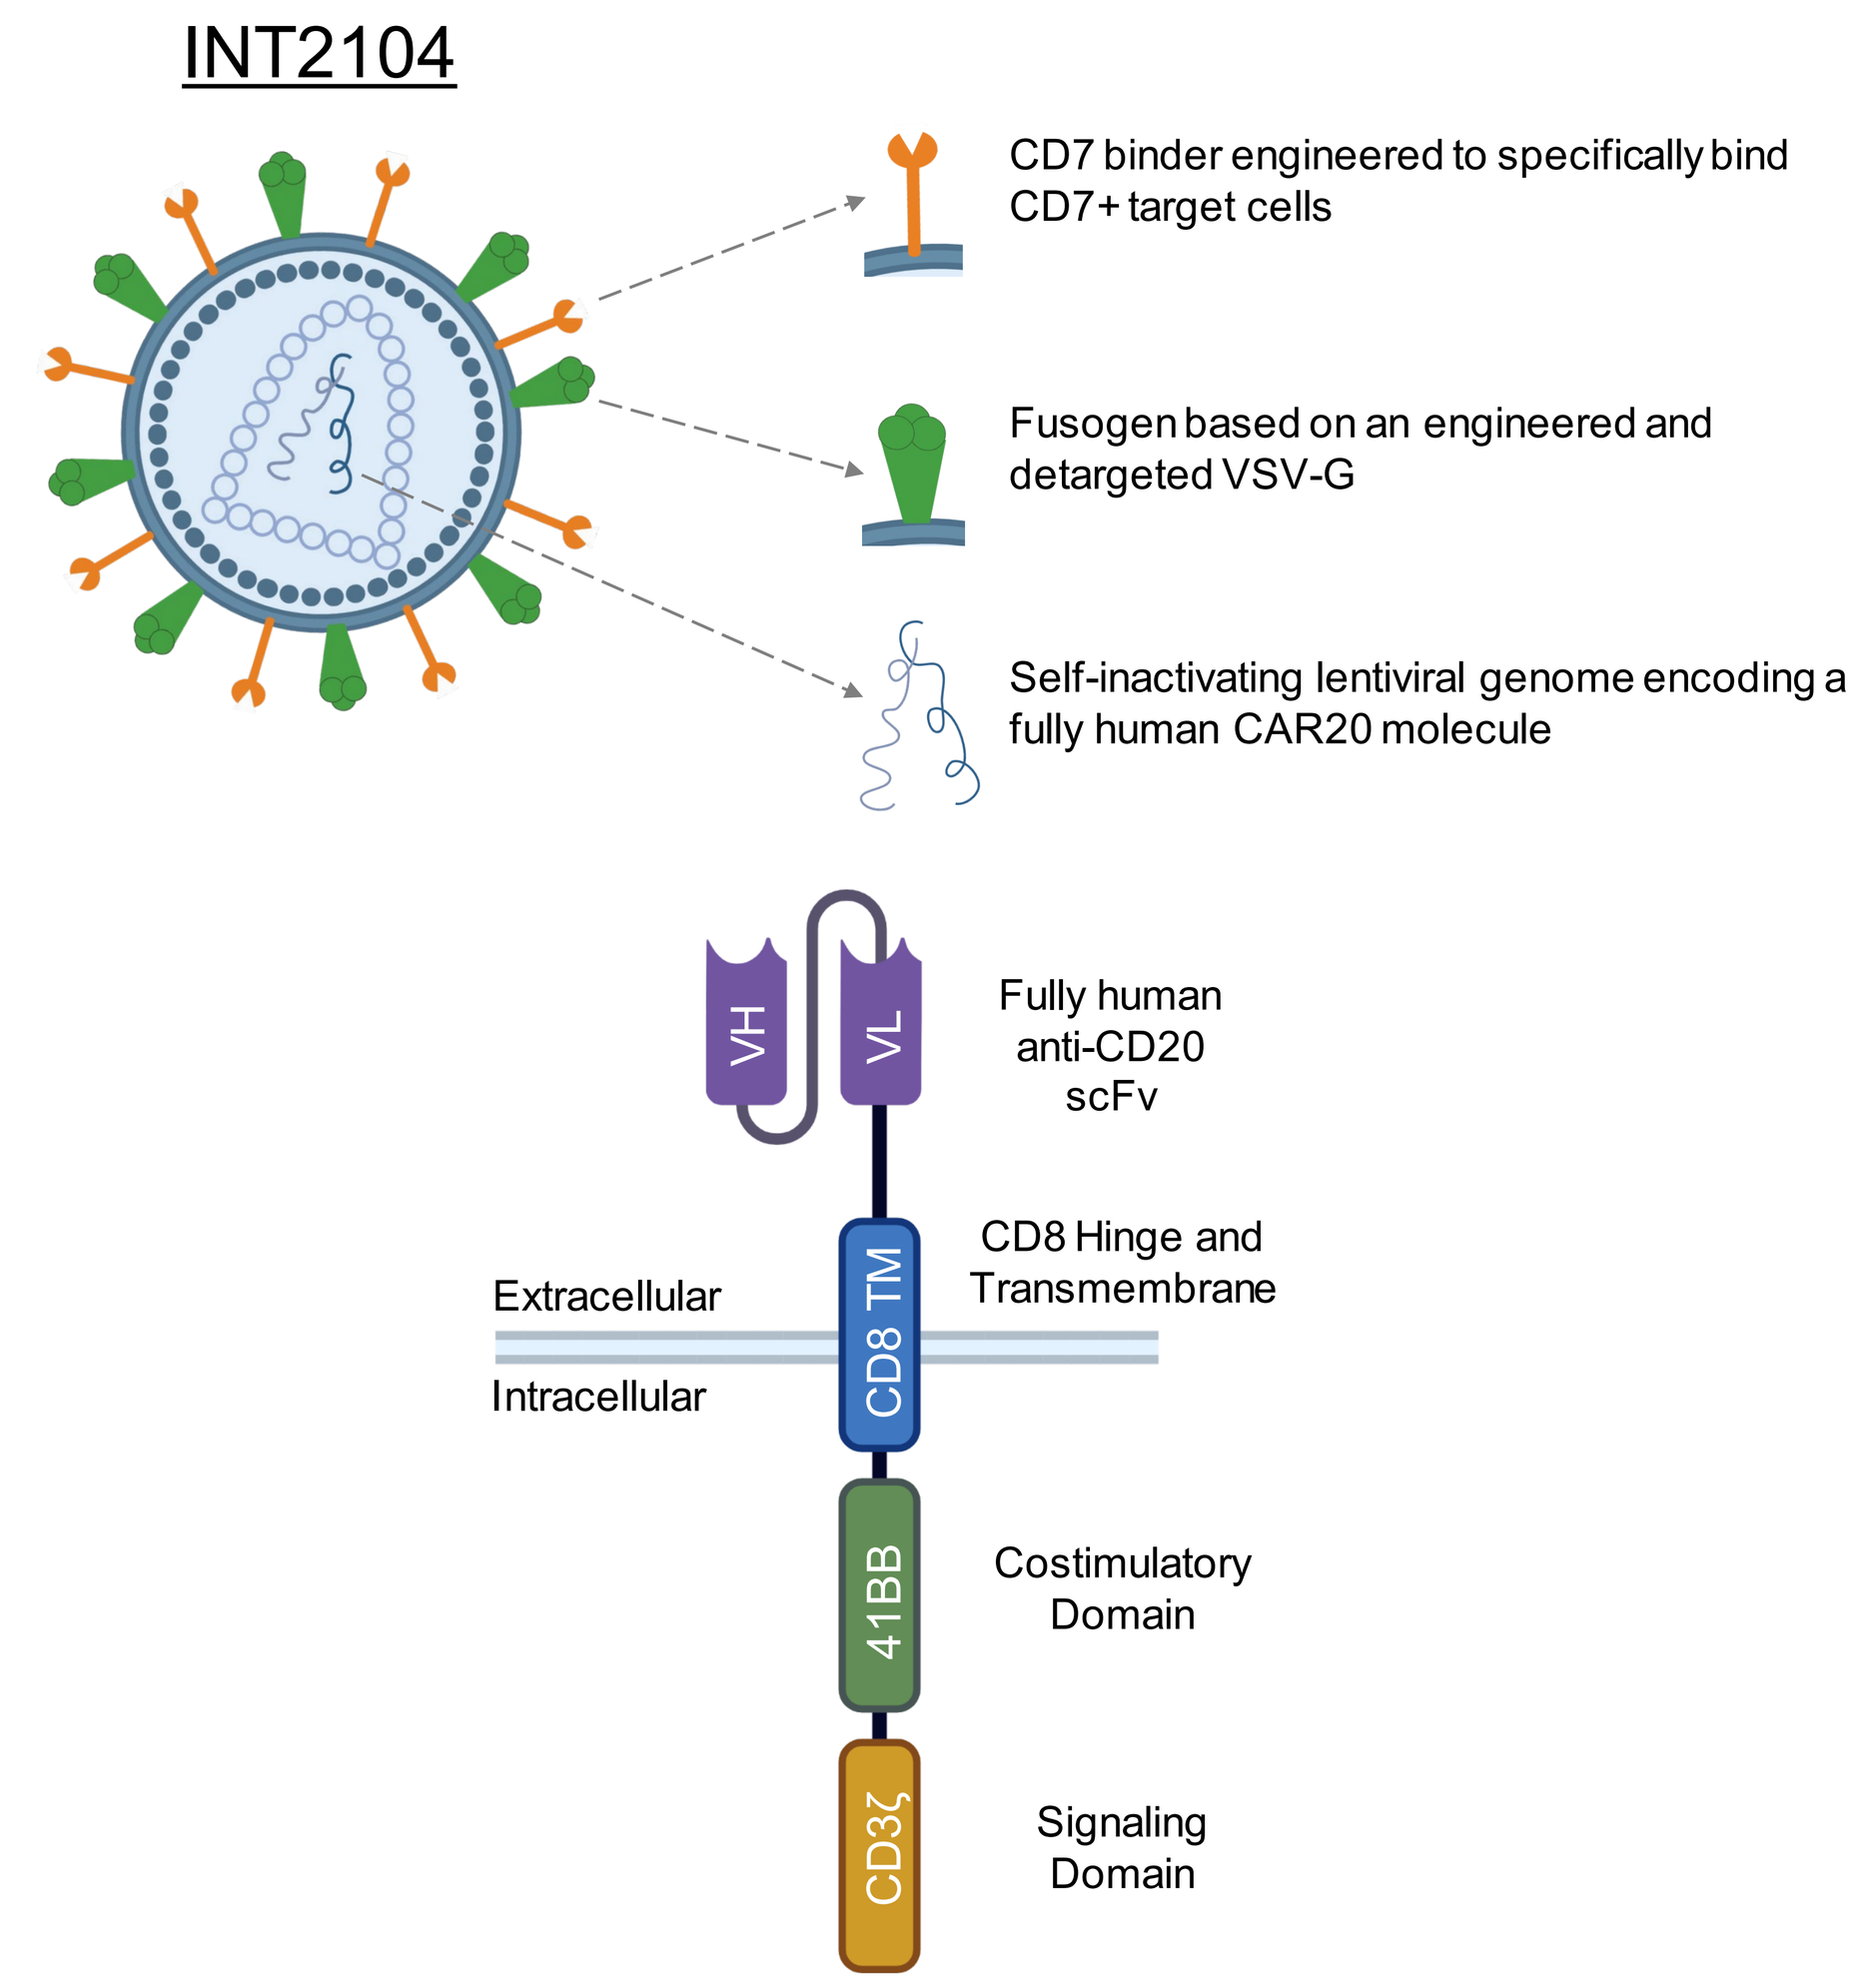
INT2104
CD7 binder engineered to specifically bind CD7+ target cells
Fusogen based on an engineered and detargeted VSV-G
Self-inactivating lentiviral genome encoding a fully human CAR20 molecule
VH
VL
Fully human anti-CD20 scFv
CD8 TM
CD8 Hinge and Transmembrane
Extracellular
Intracellular
41BB
Costimulatory Domain
CD3ζ
Signaling Domain

Rationally designed vector components

- Targeted delivery of INT2104 is enabled by engineered fusogen and binder components incorporated within the lentiviral vector
- Proprietary fusogen mutated to blind native binding of vesicular stomatitis virus G (VSV-G) to the LDL receptor while maintaining pH-dependent fusion
- CD7 Binder incorporates a proprietary human and NHP cross-reactive scFv with a human IgG1-based stalk to specifically target particles to CD7+ cells

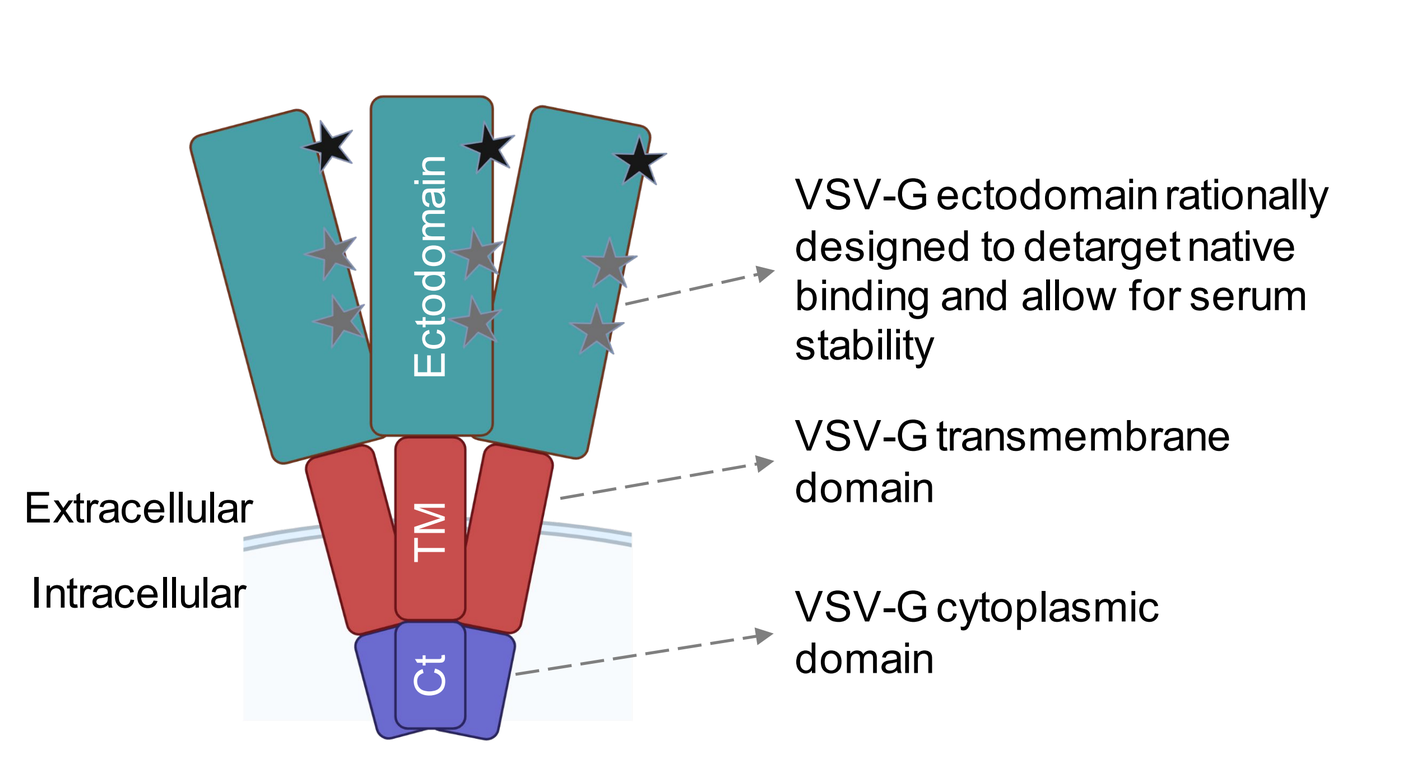
Fusogen
Ectodomain
TM
Ct
Extracellular
Intracellular
VSV-G ectodomain rationally designed to detarget native binding and allow for serum stability
VSV-G transmembrane domain
VSV-G cytoplasmic domain

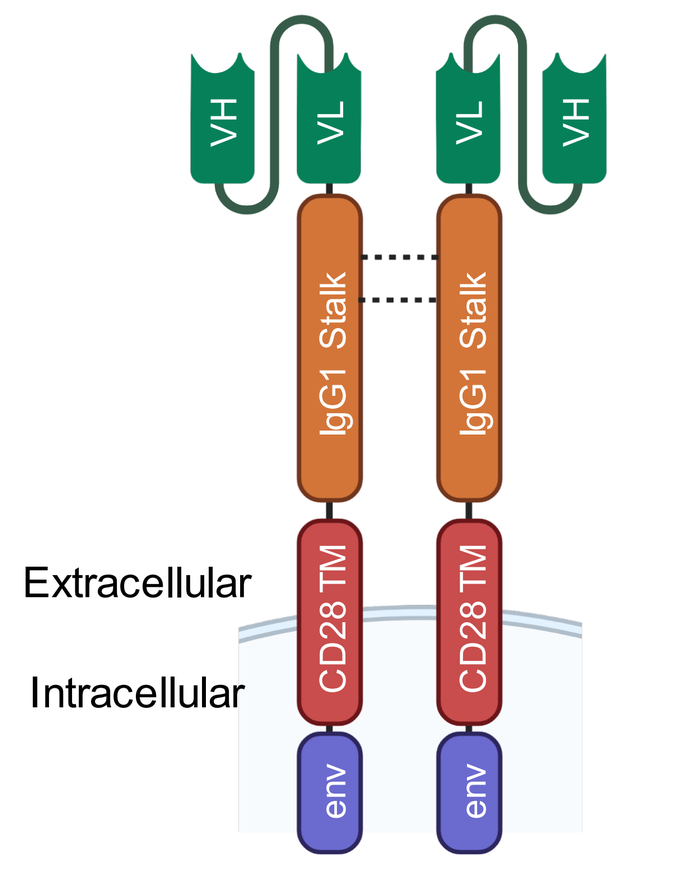
CD7 Binder
VH
VL
IgG1 Stalk
CD28 TM
env
Extracellular
Intracellular

Specific transduction of $CD7^+$ cells

- *In vitro* screening of INT2104 with GFP-encoding vectors revealed dose-dependent transduction of $CD7^+$ target cells
- No transduction evident when adding INT2104 to $CD7^-$ B cell lines

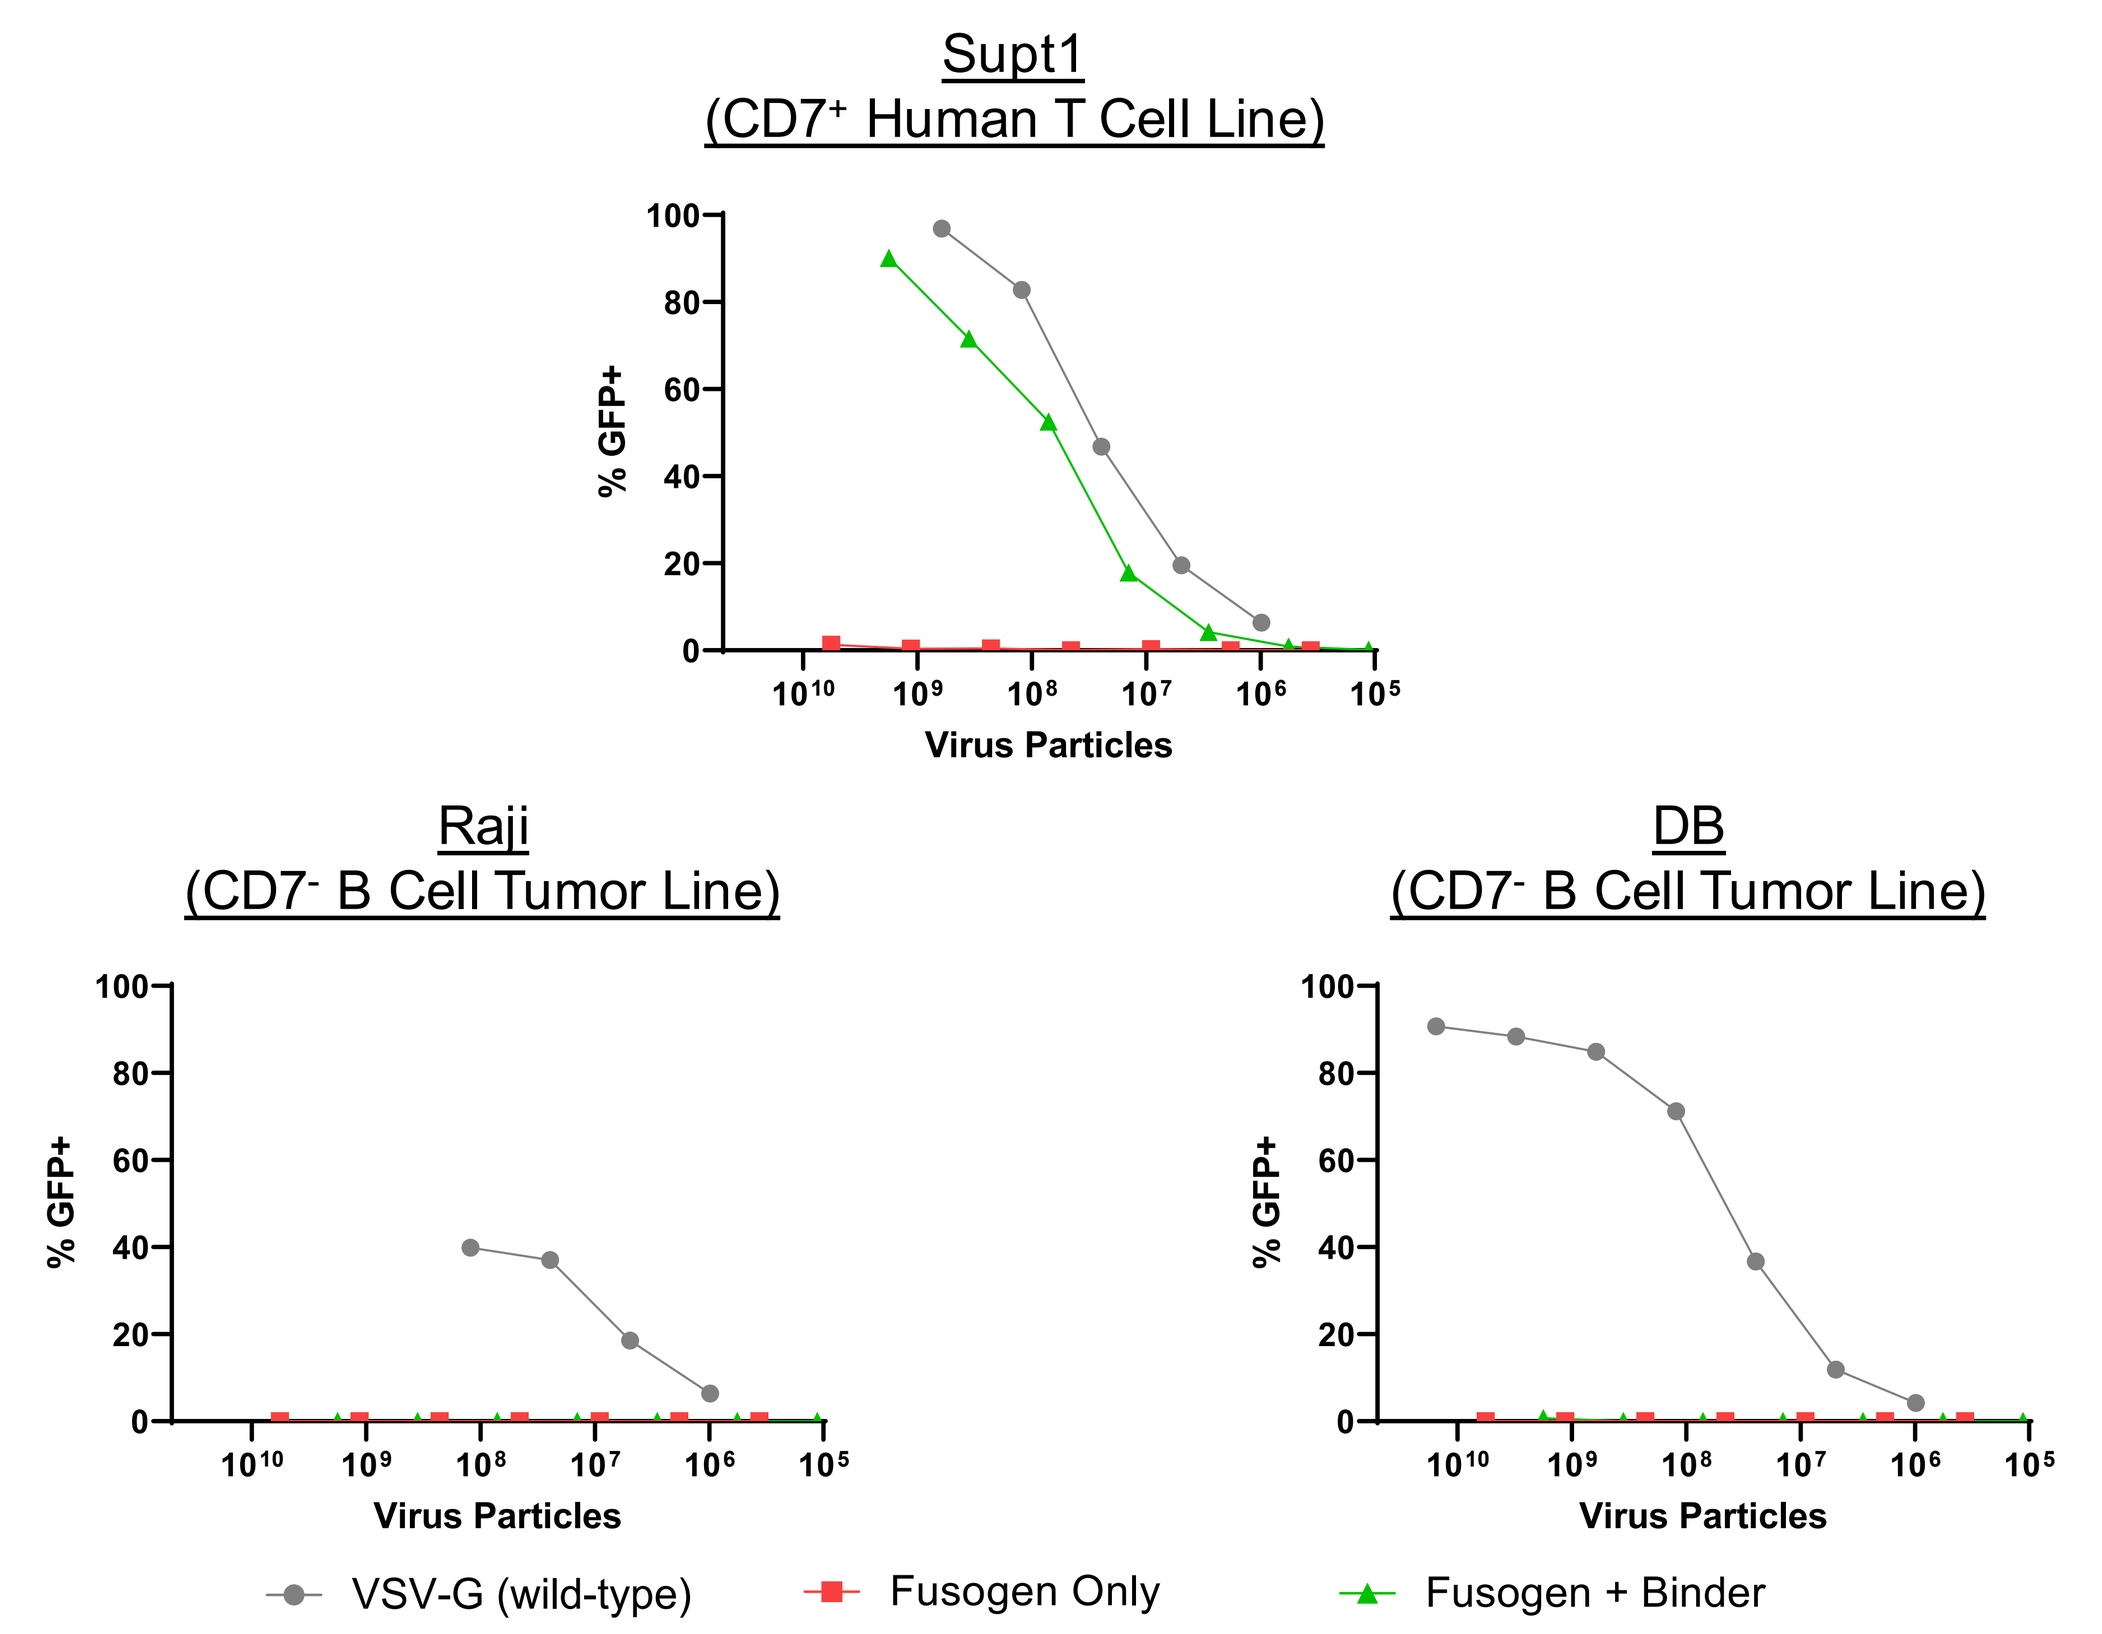
Supt1
(CD7+ Human T Cell Line)
Raji
(CD7- B Cell Tumor Line)
DB
(CD7- B Cell Tumor Line)
% GFP+
Virus Particles
VSV-G (wild-type)
Fusogen Only
Fusogen + Binder

CD7 distribution in primary human PBMCs

- Screening of PBMCs from ten unique human donors reveals T cells (both $CD4^+$ and $CD8^+$) and NK cells are the majority of cells expressing CD7 in blood
- These cells are the potential targets of INT2104 and are intended to provide the therapeutic benefit

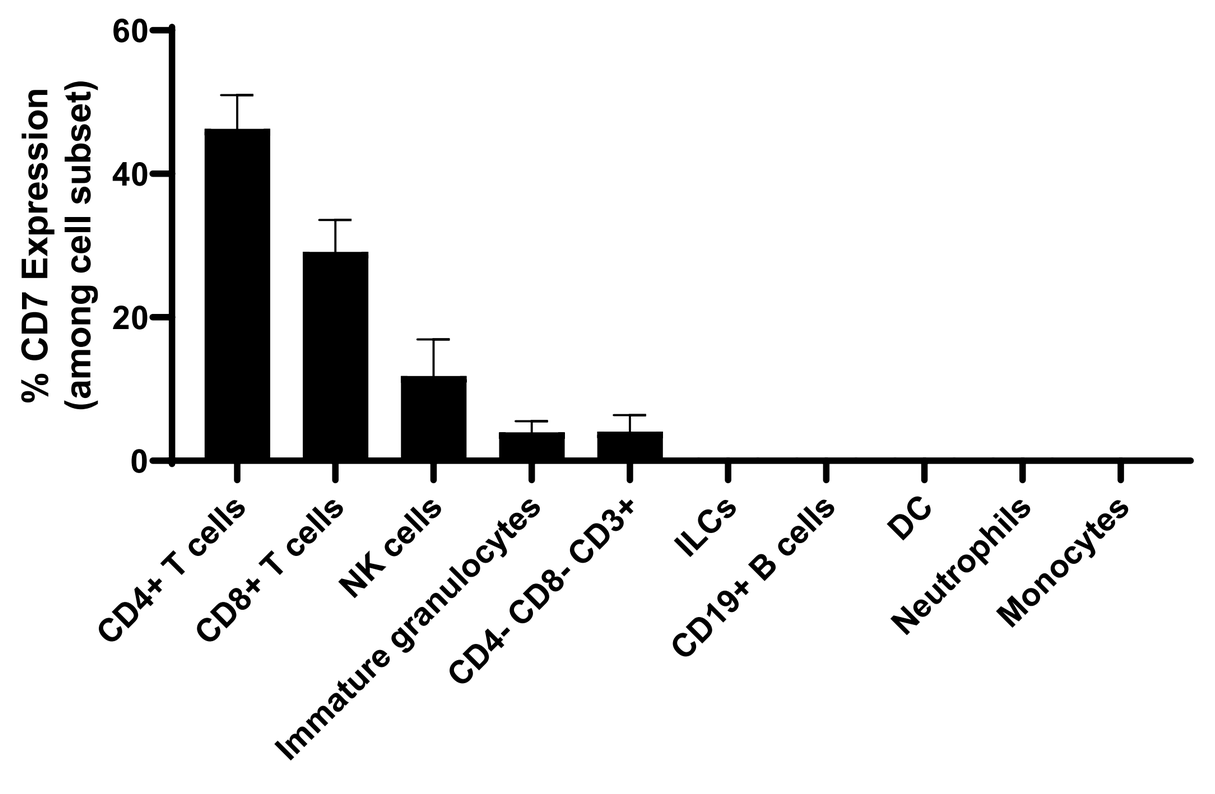
% CD7 Expression (among cell subset)
CD4+ T cells
CD8+ T cells
NK cells
Immature granulocytes
CD4- CD8- CD3+
ILCs
CD19+ B cells
DC
Neutrophils
Monocytes

Transduction of both T cells and NK cells

- Inoculation of activated primary human PBMCs with GFP-encoding vectors results in transduction of intended cell targets

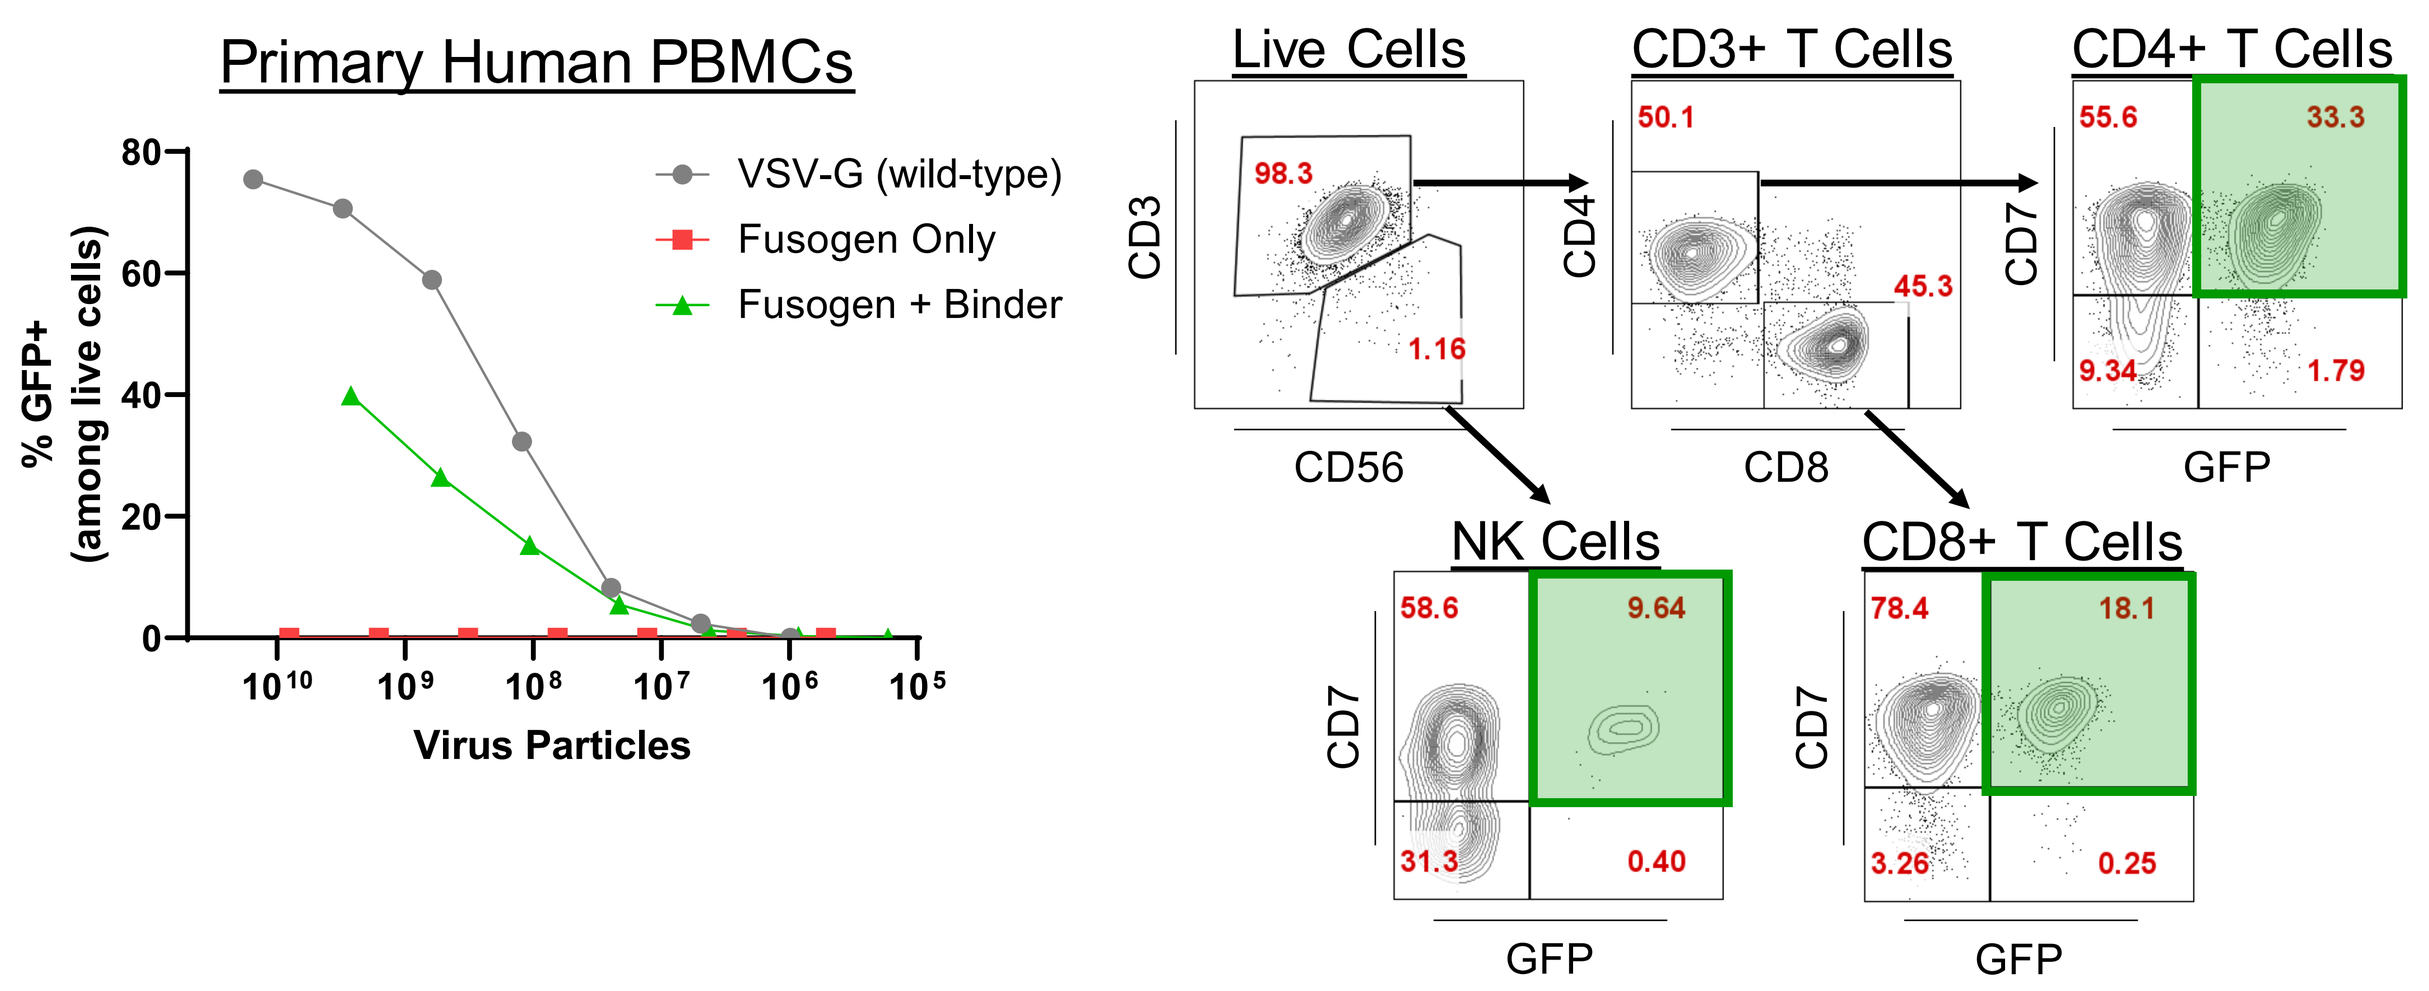
Primary Human PBMCs
% GFP+ (among live cells)
Virus Particles
VSV-G (wild-type)
Fusogen Only
Fusogen + Binder
Live Cells
CD3+ T Cells
CD4+ T Cells
NK Cells
CD8+ T Cells
CD3
CD56
CD4
CD8
CD7
GFP
98.3
1.16
50.1
45.3
55.6
33.3
9.34
1.79
58.6
9.64
31.3
0.40
78.4
18.1
3.26
0.25

CAR mediated, dose-dependent B cell killing

- Incubation of activated primary human PBMCs with INT2104 generates $CD4^+$ T cells, $CD8^+$ T cells, and NK cells that are CAR^+
- CAR^+ cells co-cultured with B cell tumor targets show dose-dependent killing

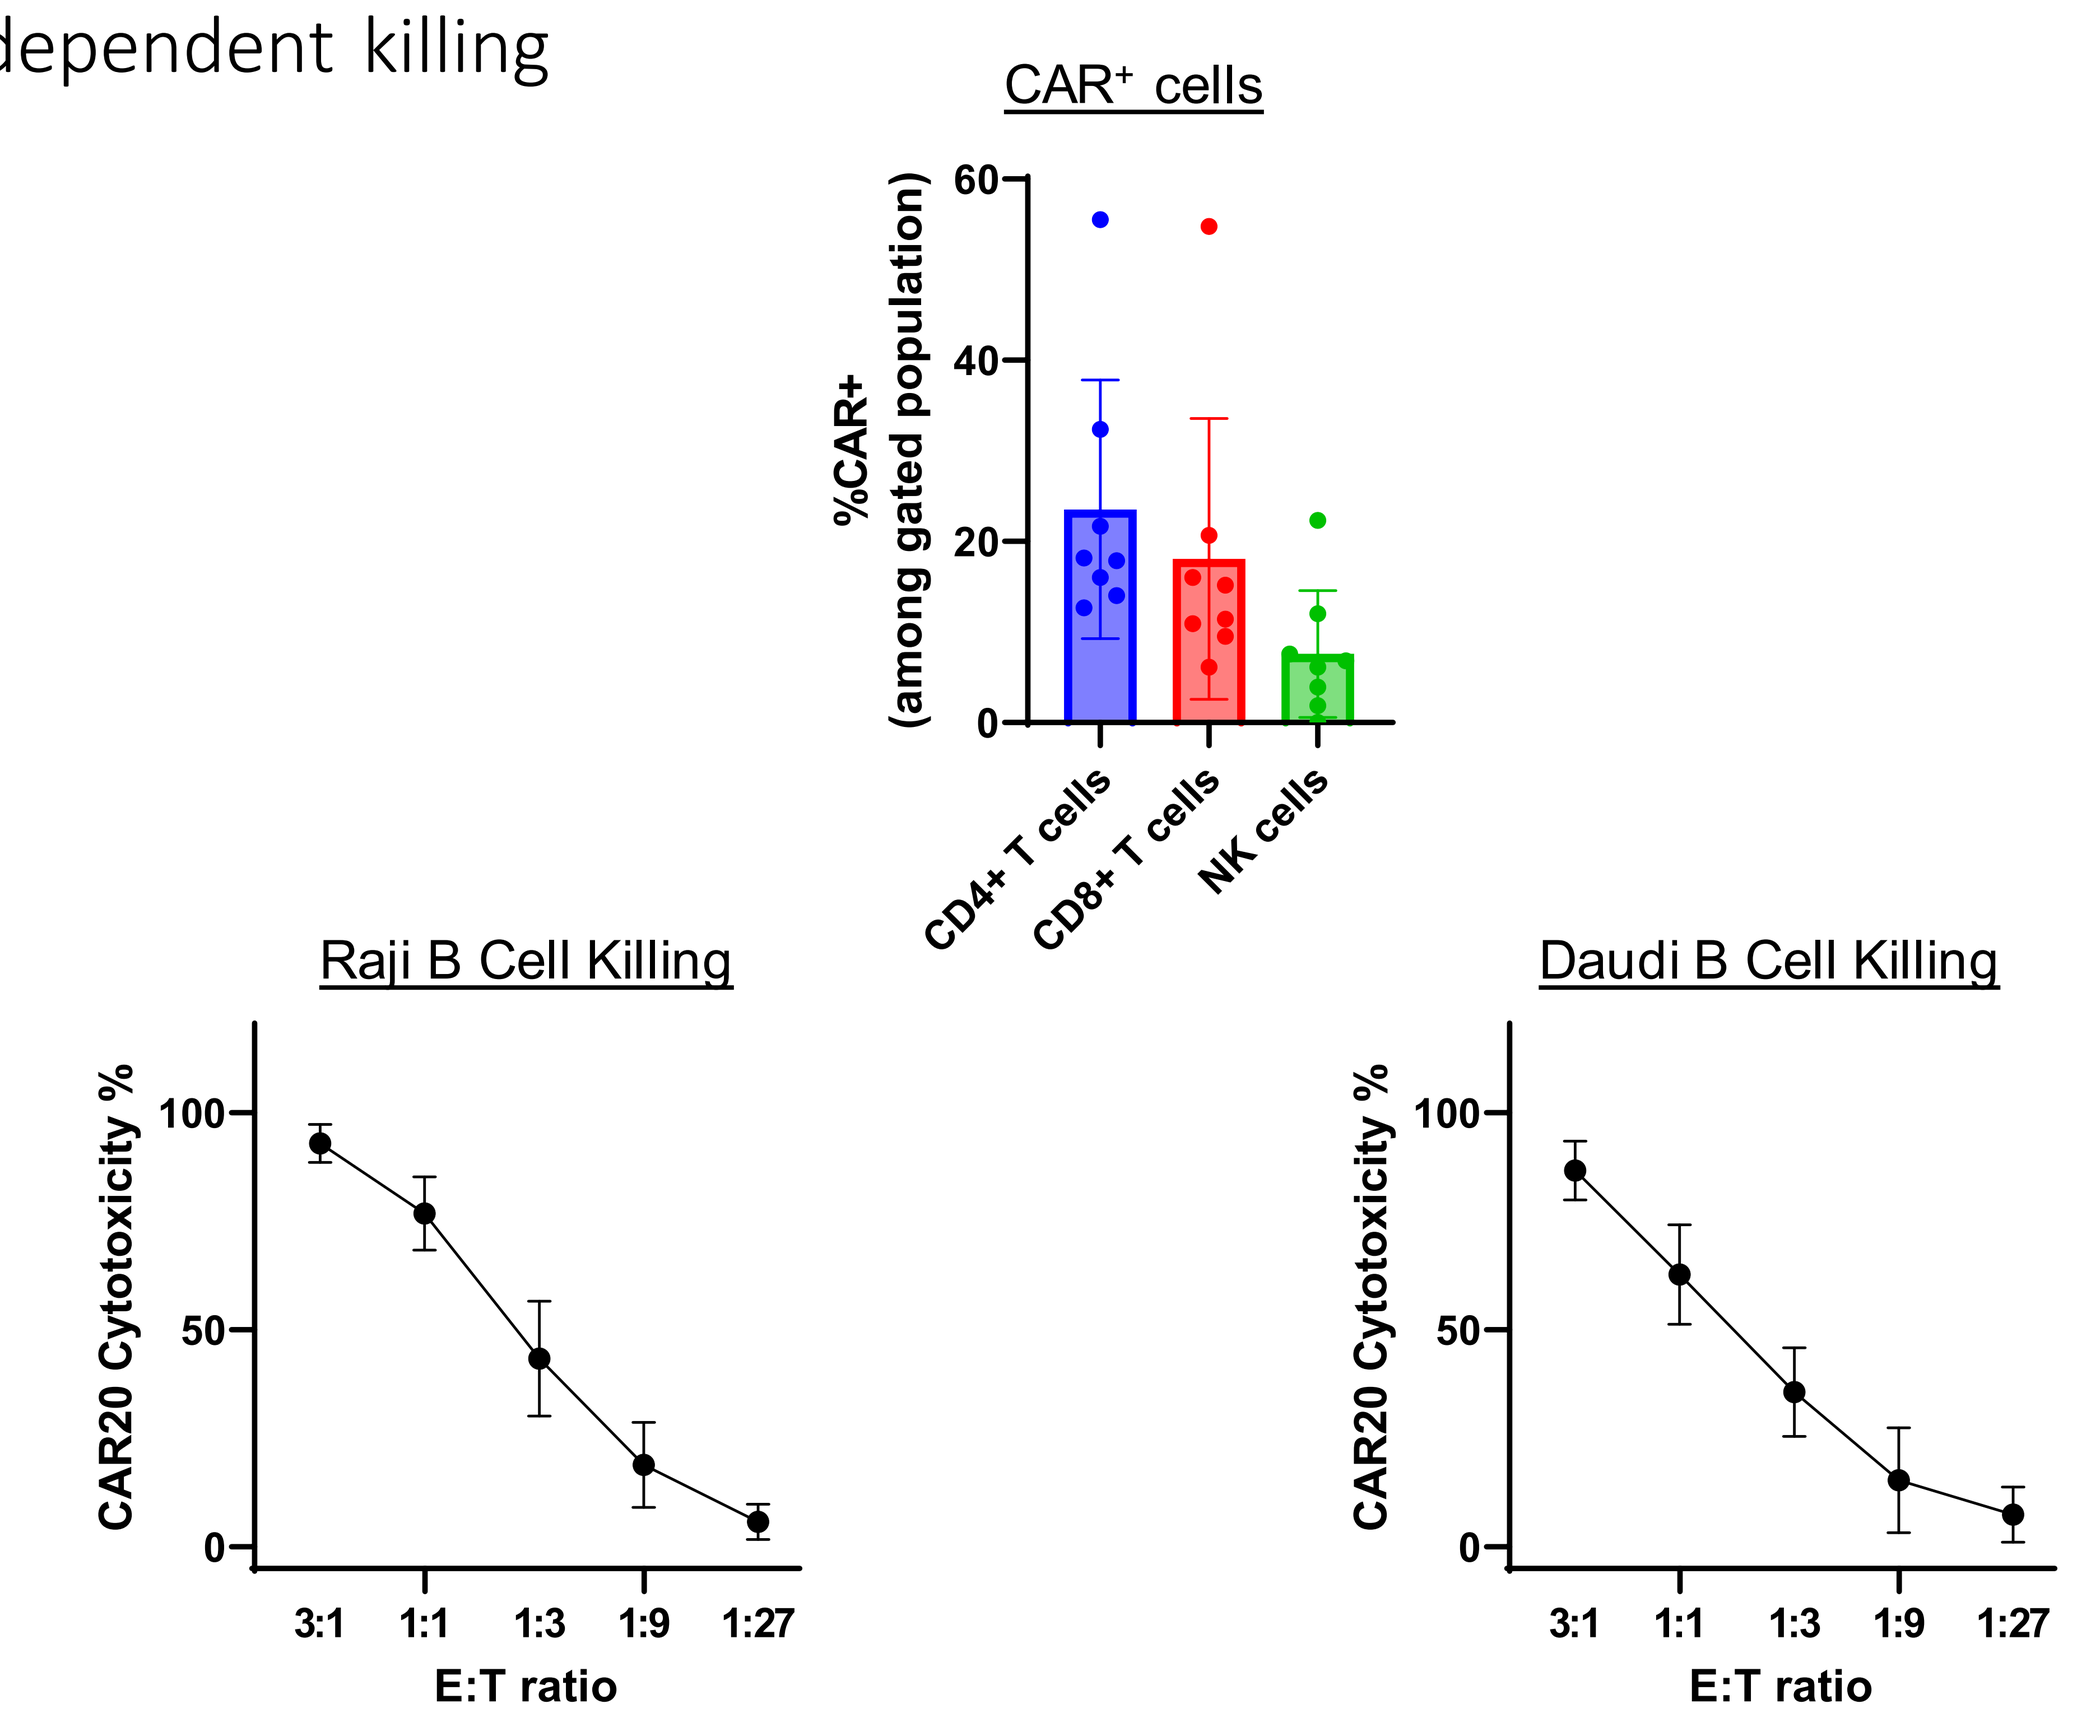
CAR+ cells
%CAR+ (among gated population)
CD4+ T cells
CD8+ T cells
NK cells
Raji B Cell Killing
Daudi B Cell Killing
CAR20 Cytotoxicity %
3:1
1:1
1:3
1:9
1:27
E:T ratio

No B cell transduction detected following INT2104 treatment

- Further assessment into potential off-target transduction of B cells was conducted with six B cell tumor lines and primary PBMCs isolated from patients with B cell malignancies which were inoculated with INT2104
- No evidence of B cell transduction was seen across a wide range of MOIs

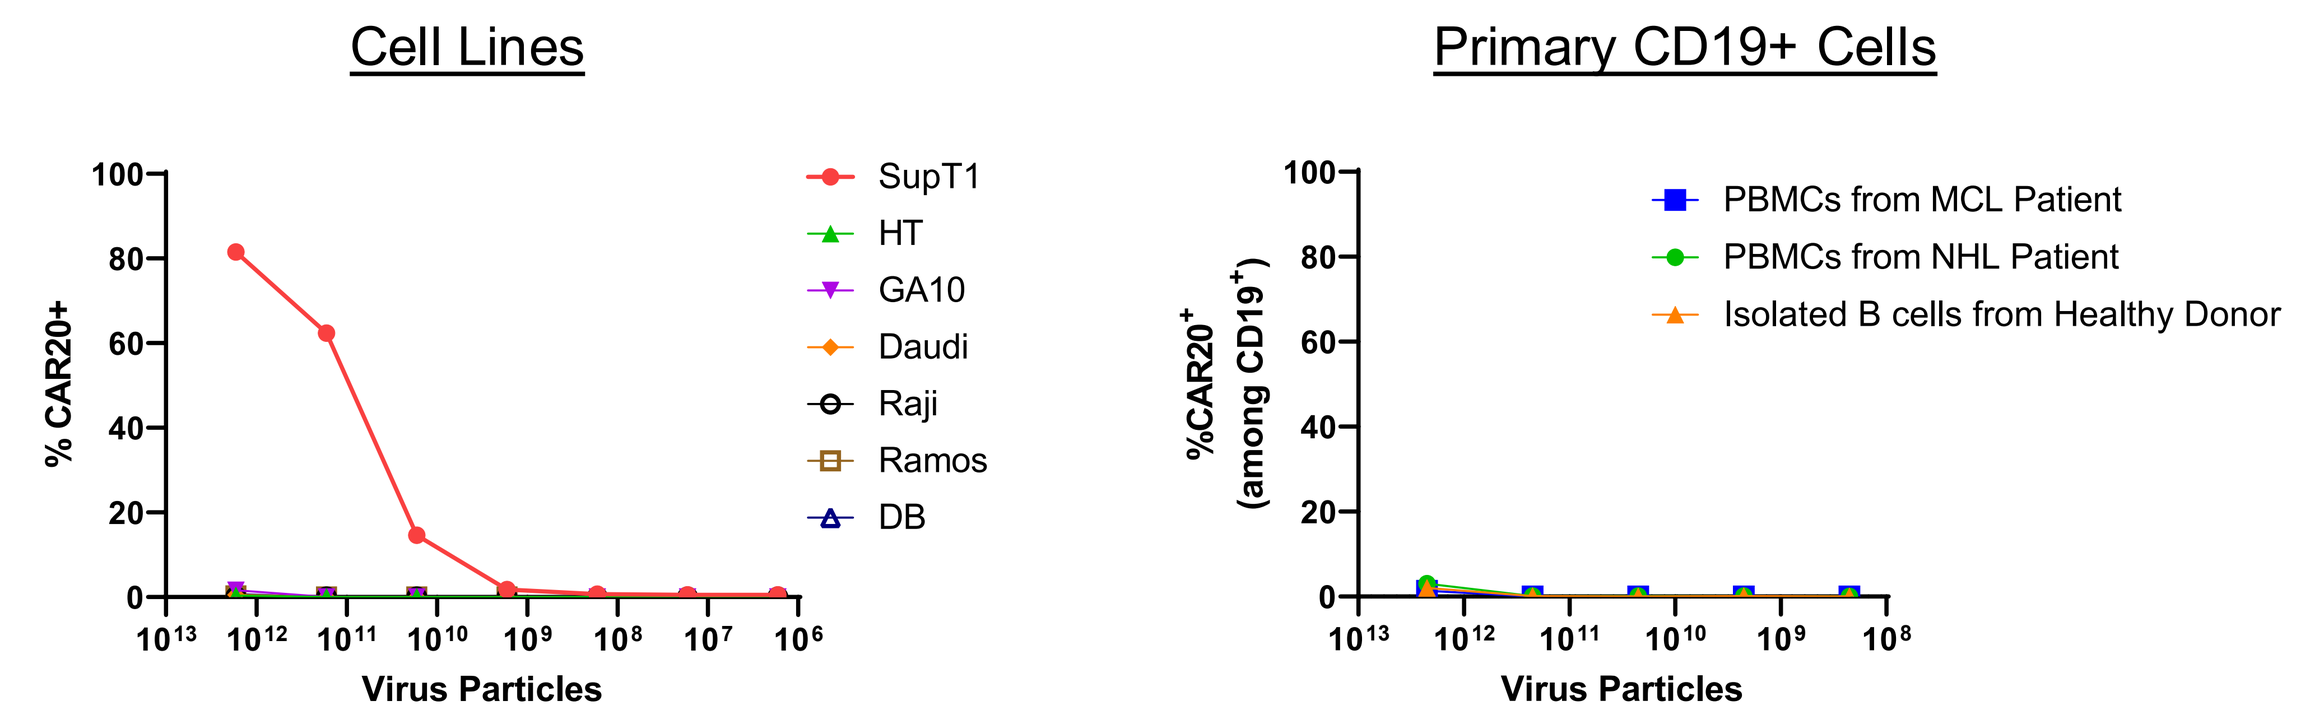
Cell Lines
% CAR20+
Virus Particles
SupT1
HT
GA10
Daudi
Raji
Ramos
DB
Primary CD19+ Cells
%CAR20+ (among CD19+)
PBMCs from MCL Patient
PBMCs from NHL Patient
Isolated B cells from Healthy Donor

Killing of CAR^+ B cells with INT2104-treated cells

- An experiment was performed to assess the risk of unintended B cell transduction resulting in CAR-resistant B cell tumors
- INT2104-transduced PBMCs killed B cells transduced with CAR20 and GFP similarly
- No evidence of selective advantage towards transduced B cells was seen

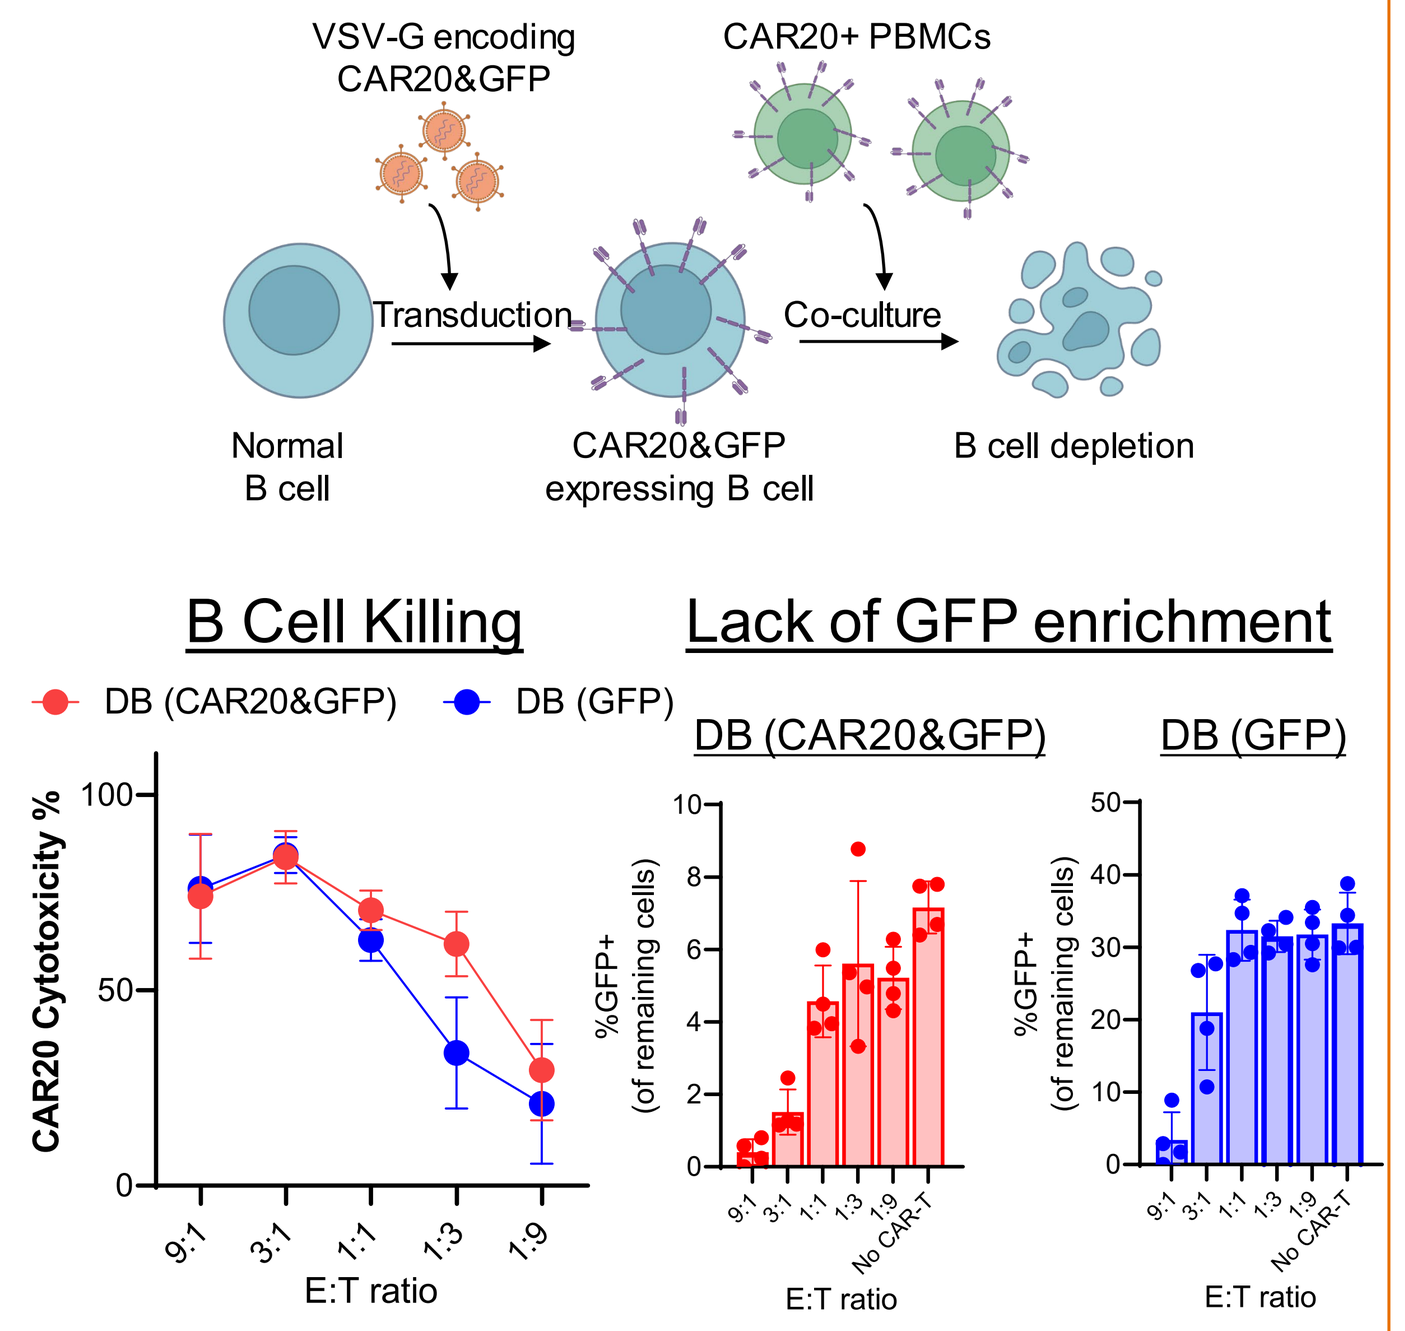
VSV-G encoding CAR20&GFP
CAR20+ PBMCs
Transduction
Co-culture
Normal B cell
CAR20&GFP expressing B cell
B cell depletion
B Cell Killing
DB (CAR20&GFP)
DB (GFP)
CAR20 Cytotoxicity %
E:T ratio
Lack of GFP enrichment
DB (CAR20&GFP)
DB (GFP)
%GFP+ (of remaining cells)
No CAR-T

Endogenous B cell depletion after *in vivo* delivery

- Intravenous delivery of INT2104 to $CD34^+$ engrafted NSG mice results in B cell depletion
- CAR^+ cells are detectable in peripheral blood starting at Day 7 post-injection and throughout the remainder of the study

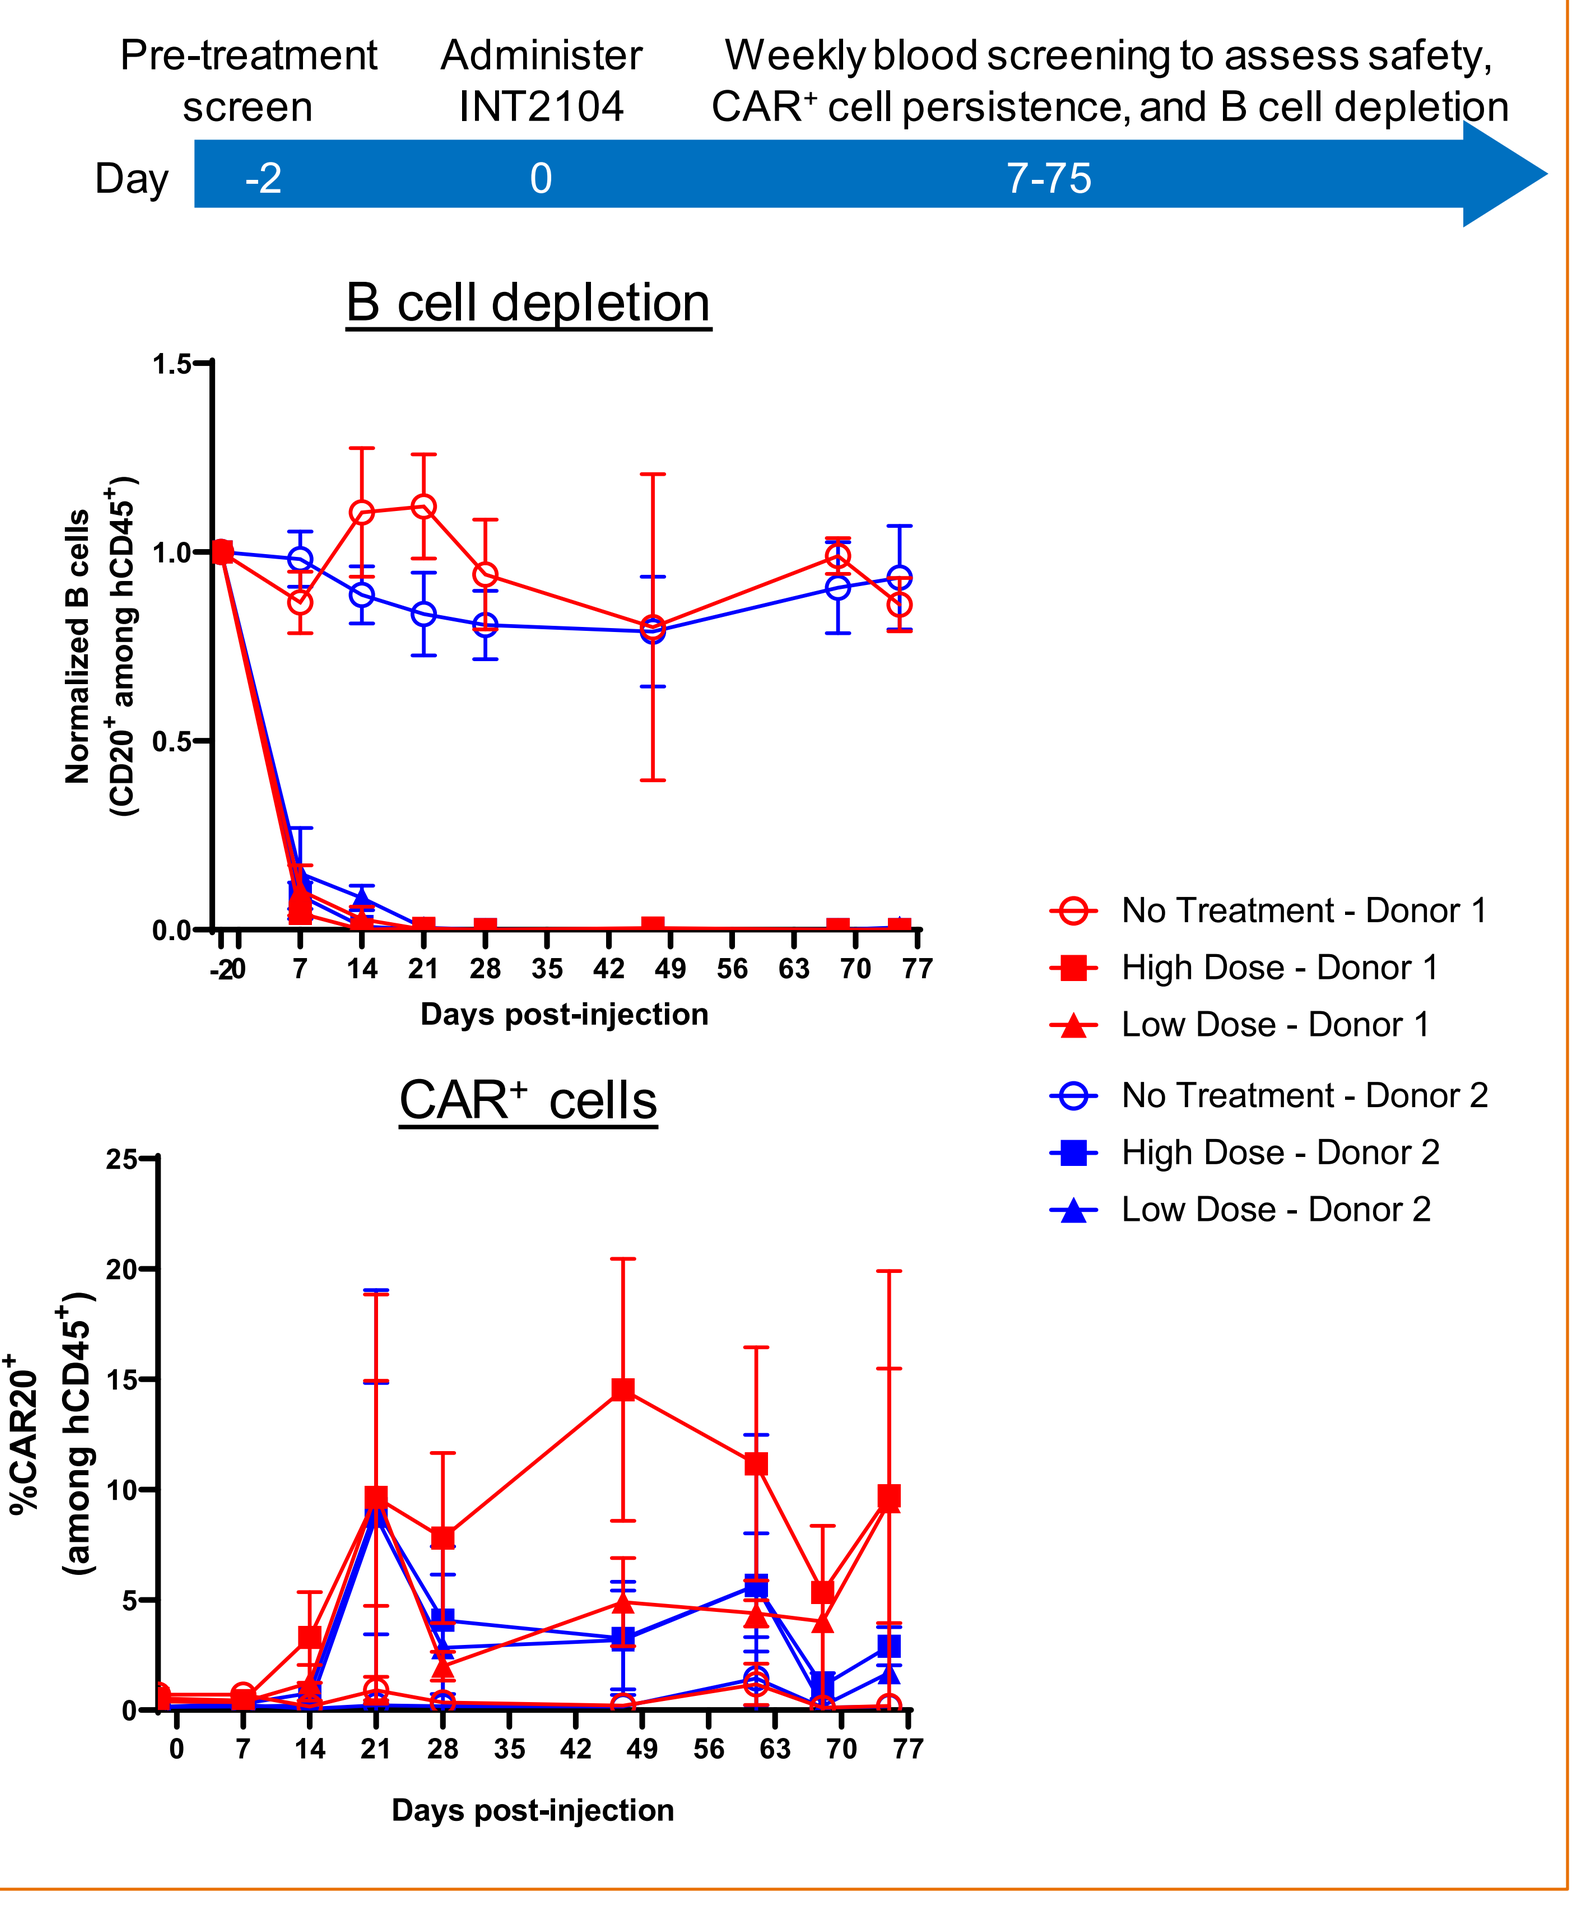
Pre-treatment screen
Administer INT2104
Weekly blood screening to assess safety, CAR+ cell persistence, and B cell depletion
Day
-2
0
7-75
B cell depletion
Normalized B cells (CD20+ among hCD45+)
Days post-injection
No Treatment - Donor 1
High Dose - Donor 1
Low Dose - Donor 1
No Treatment - Donor 2
High Dose - Donor 2
Low Dose - Donor 2
CAR+ cells
%CAR20+ (among hCD45+)

Ablation of established B cell tumors

- Established Raji B cell tumors can be controlled following INT2104 administration to NSG MHCI/II KO mice

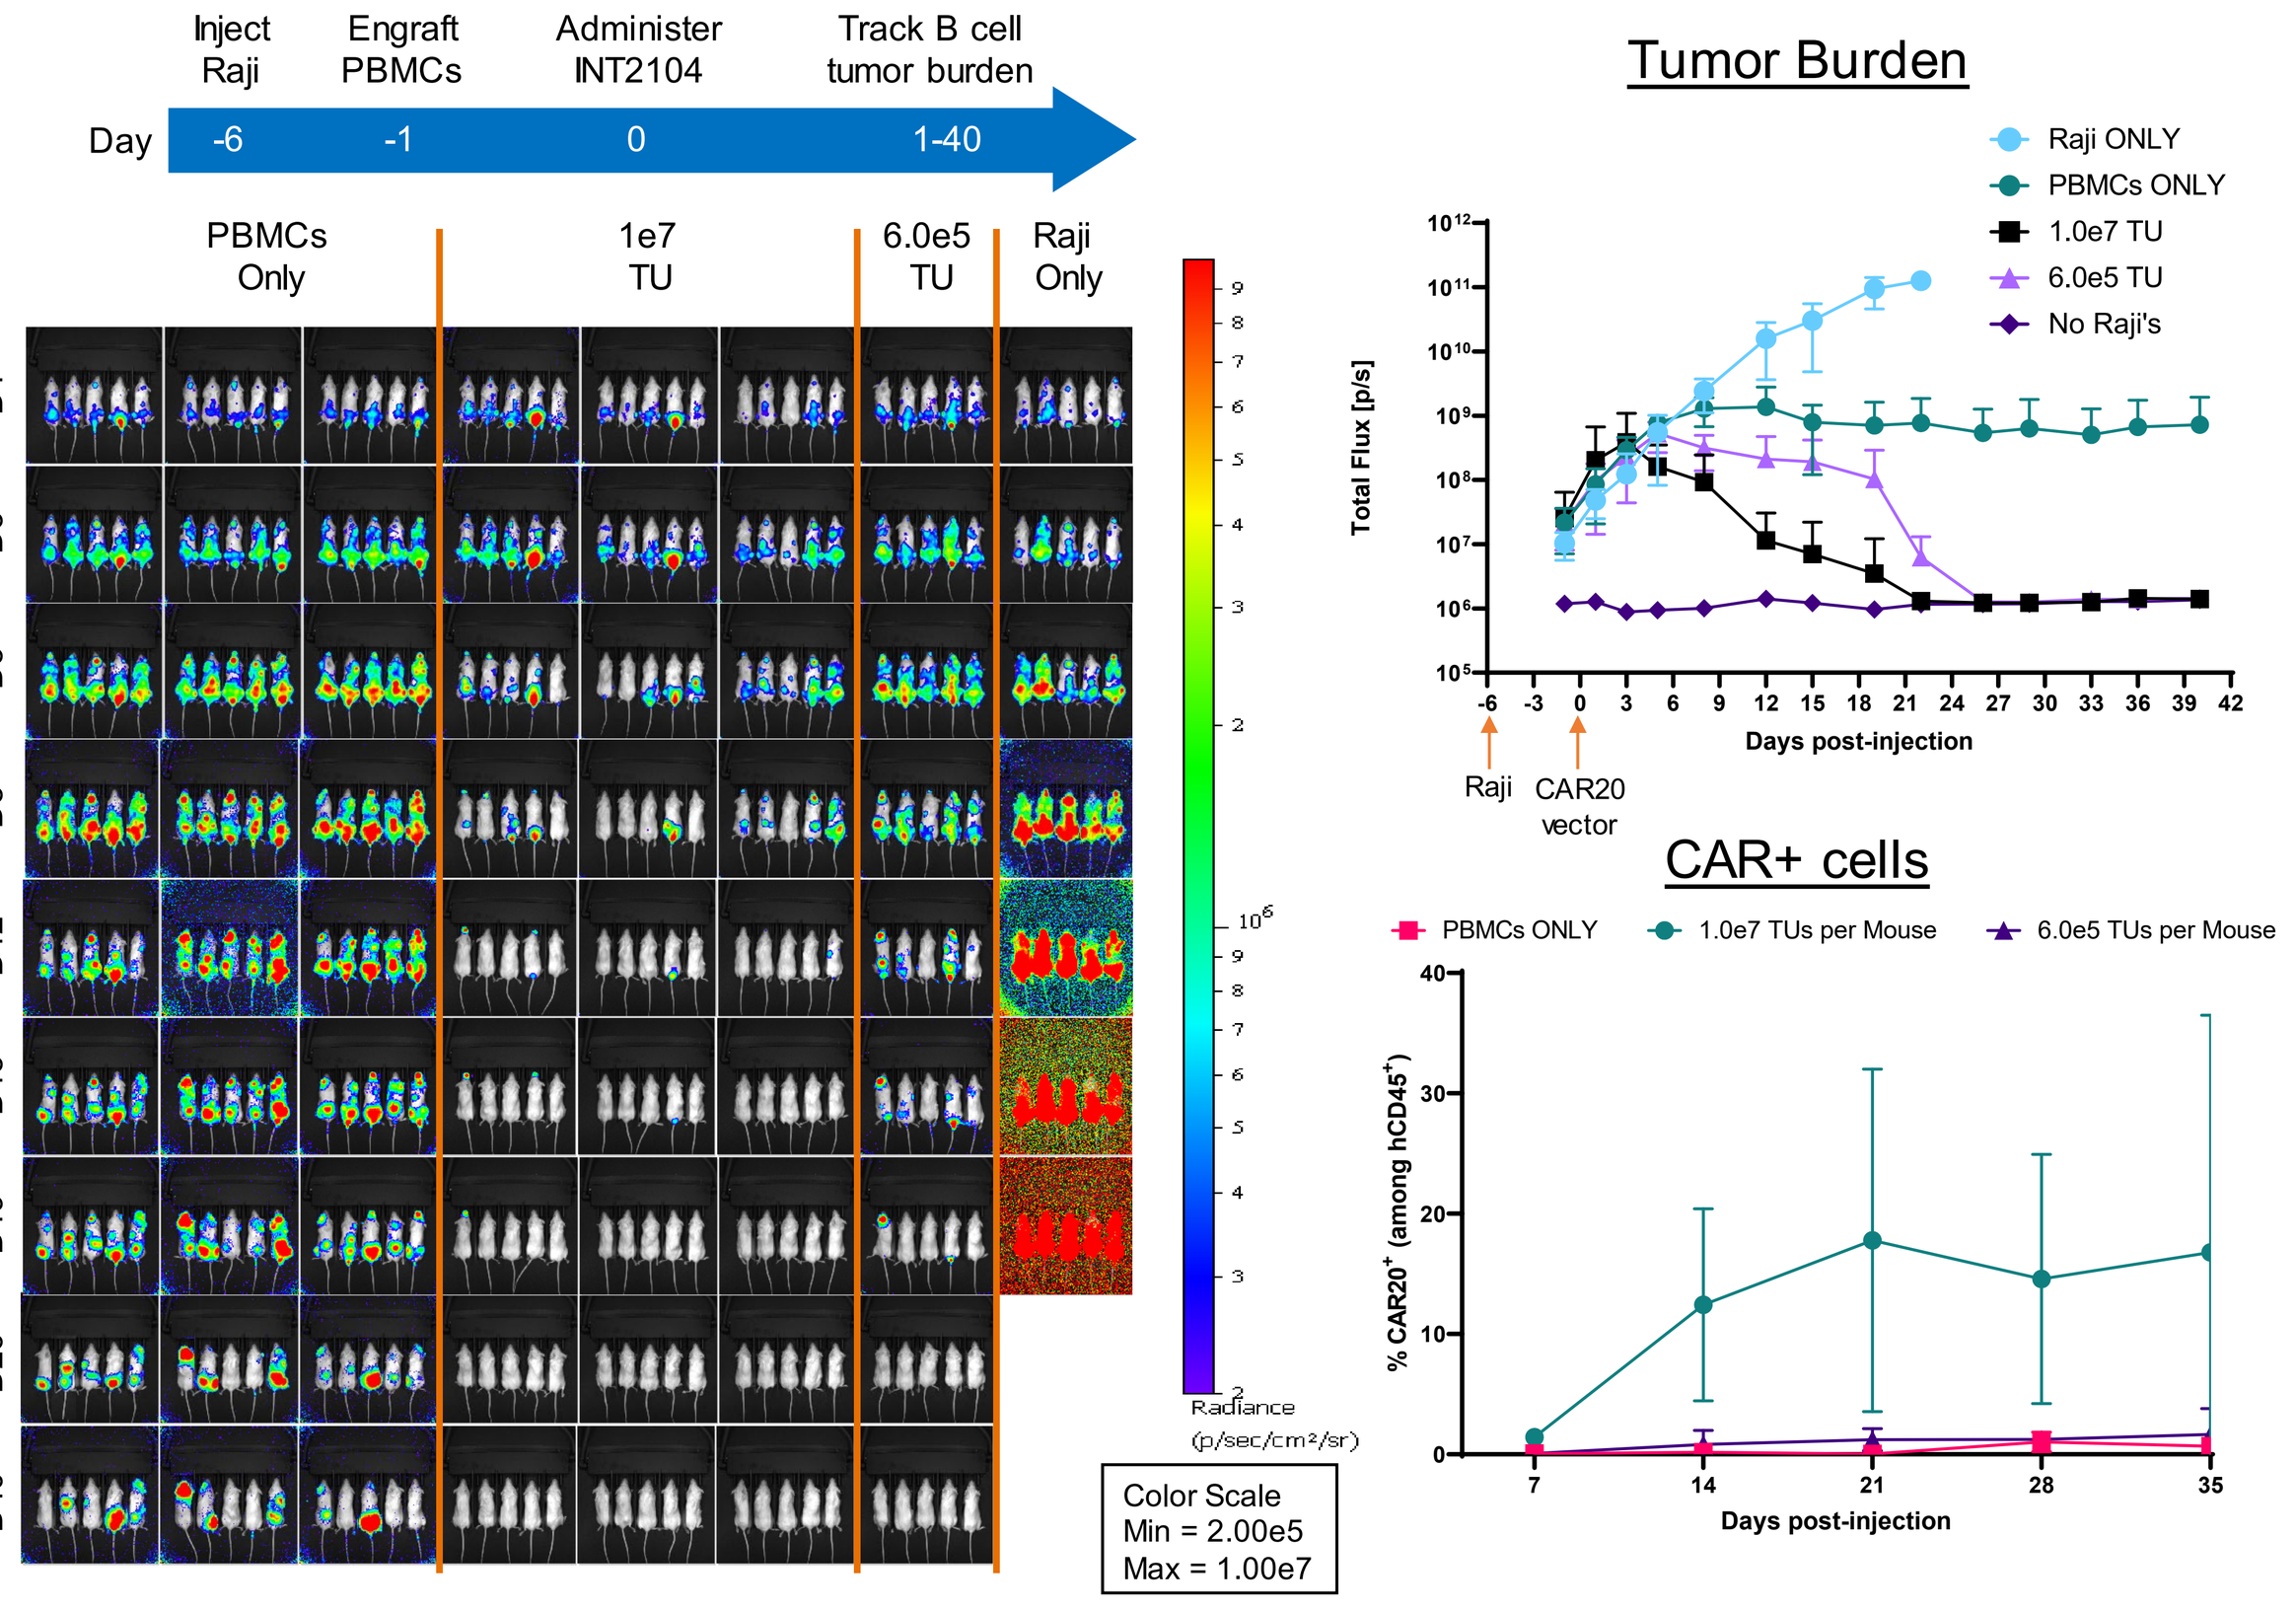
Inject Raji
Engraft PBMCs
Administer INT2104
Track B cell tumor burden
Day
-6
-1
0
1-40
PBMCs Only
1e7 TU
6.0e5 TU
Raji Only
D1
D3
D5
D8
D12
D15
D19
D26
D40
Radiance (p/sec/cm²/sr)
Color Scale
Min = 2.00e5
Max = 1.00e7
Tumor Burden
Total Flux [p/s]
Days post-injection
Raji ONLY
PBMCs ONLY
1.0e7 TU
6.0e5 TU
No Raji's
Raji
CAR20 vector
CAR+ cells
PBMCs ONLY
1.0e7 TUs per Mouse
6.0e5 TUs per Mouse
% CAR20+ (among hCD45+)

Looking forward with Interius

- INT2104 exhibits specificity, biological activity, and safety following IV delivery, supporting continued development and transition into the clinic
- Plug and play nature of the Interius bioplatform enables development of new products by changing binder and/or transgene

Acknowledgements

- Thanks to Ronnie Russell, Bruce Schnepp, Jeffery Foss, Debu Boral, Ramesh Singh, Daniel Grubaugh, Julia Nguyen, and Leticia Kuri-Cervantes for designing/performing these studies
- Thanks to the AD, PD, and DPD teams for providing vector to support these studies

© Interius BioTherapeutics, Inc. 2023. All rights reserved. Some images have been generated using BioRender.com. Dr. Saar Gill is a co-founder, collaborator, consultant, and equity-holder of Interius BioTherapeutics, Inc. Contact: info@Interiusbio.com or Interiusbio.com.